BIOAVAILABILITY OF EPICATECHIN AFTER CONSUMPTION OF GRAPE SEED EXTRACT IN HUMANS

Natalie Eich[1], Erik Schneider[1], John Cuomo[1], Alexander Rabovsky[1], Joseph A. Vita[2], Joseph Palmisano[2], Monika Holbrook[2]

[1]Department of Research & Development, USANA Health Sciences, Salt Lake City, UT, USA [2]Boston University School of Medicine

INTRODUCTION

Diets that are rich in plant foods have been associated with a decreased risk for specific disease processes and certain chronic diseases. In addition to essential macronutrients and micronutrients, the flavonoids in a variety of plant foods may have health-enhancing properties. Grape seed extract (GSE) is a common dietary supplement that is known to be rich in the flavan-3-ols, commonly known as catechins. However, the bioavailability and the biological effects of the grape seed extract flavonoids are poorly understood. To begin to address these issues, we developed a method based on liquid chromatography-mass spectrometry (LC-MS) detection to determine physiological levels of epicatechin (EC).

Specifically, we evaluated the appearance of epicatechin after supplementation of 1g GSE taken orally in tablet form. A four hour curve of plasma epicatechin was obtained as measured by LC-MS. Furthermore, to correlate circulating epicatechin and antioxidant protection, we went on to determine plasma antioxidant reserve, a measure of resistance to plasma oxidative stress[2].

TABLE 1. CLINICAL BASELINE CHARACTERISTICS

CHARACTERISTIC	MEAN OR NUMBER	RANGE
Female gender (%)	4 (25%)	-
Coronary artery disease	16 (100%)	-
History of hypertension (%)	11 (69%)	-
History of diabetes mellitus (%)	6 (38%)	-
Age, years	60 ± 16	44 - 73
Fasting glucose, mg/dL	92 ± 24	50 - 141
Total cholesterol, mg/dL	142 ± 47	87 - 285
HDL cholesterol, mg/dL	41 ± 8	32 - 58
LDL cholesterol, mg/dL	74 ± 39	34 - 200
Mean triglyceride, mg/dL	138 ± 58	40 - 242

Mean ± SD, N=16

MATERIALS AND METHODS

Sixteen patients with confirmed coronary artery disease completed this study. Exclusion criteria included pregnancy, other active major illness, and treatment with another investigational drug, antioxidant vitamins, and herbal supplements within 30 days of the start of the study. Participants were instructed to avoid grapes and grape products for one week prior to the study, to fast overnight on the day of the study, and, if applicable, to avoid smoking the morning of the study.

Upon arrival at the Boston University Vascular Research Unit, an indwelling intravenous catheter was inserted in the left arm and blood was collected for measurement of baseline epicatechin, glucose, PAR and cholesterol. After consuming a standardized snack consisting of two pieces of toast, subjects received an open label 1 g oral dose of GSE (USANA Health Sciences).

Additional blood samples were collected at one, two, three, and four hours after supplementation of GSE for measurement of plasma epicatechin levels and PAR. Blood samples were centrifuged immediately after collection and plasma was stored at -80°C until analyzed.

LC-MS Determination of Epicatechin

Instrumentation

A liquid chromatography-mass spectrometry (LC-MS) system consisting of a 1200 Series LC system with an 1100 MSD bench top mass spectrometer equipped with an electrospray (ES) source (Agilent Technologies, Palo Alto, CA) was used for this investigation. A sample of human plasma (6 µl) was injected onto a 4.6 X 250 mm id, 3.5 µm Kromasil C18 analytical column (Varian, Palo Alto, CA). The method was isocratic using a 25% methanol in water mobile phase.

Plasma Epicatechin After Grape Seed Extract Supplementation

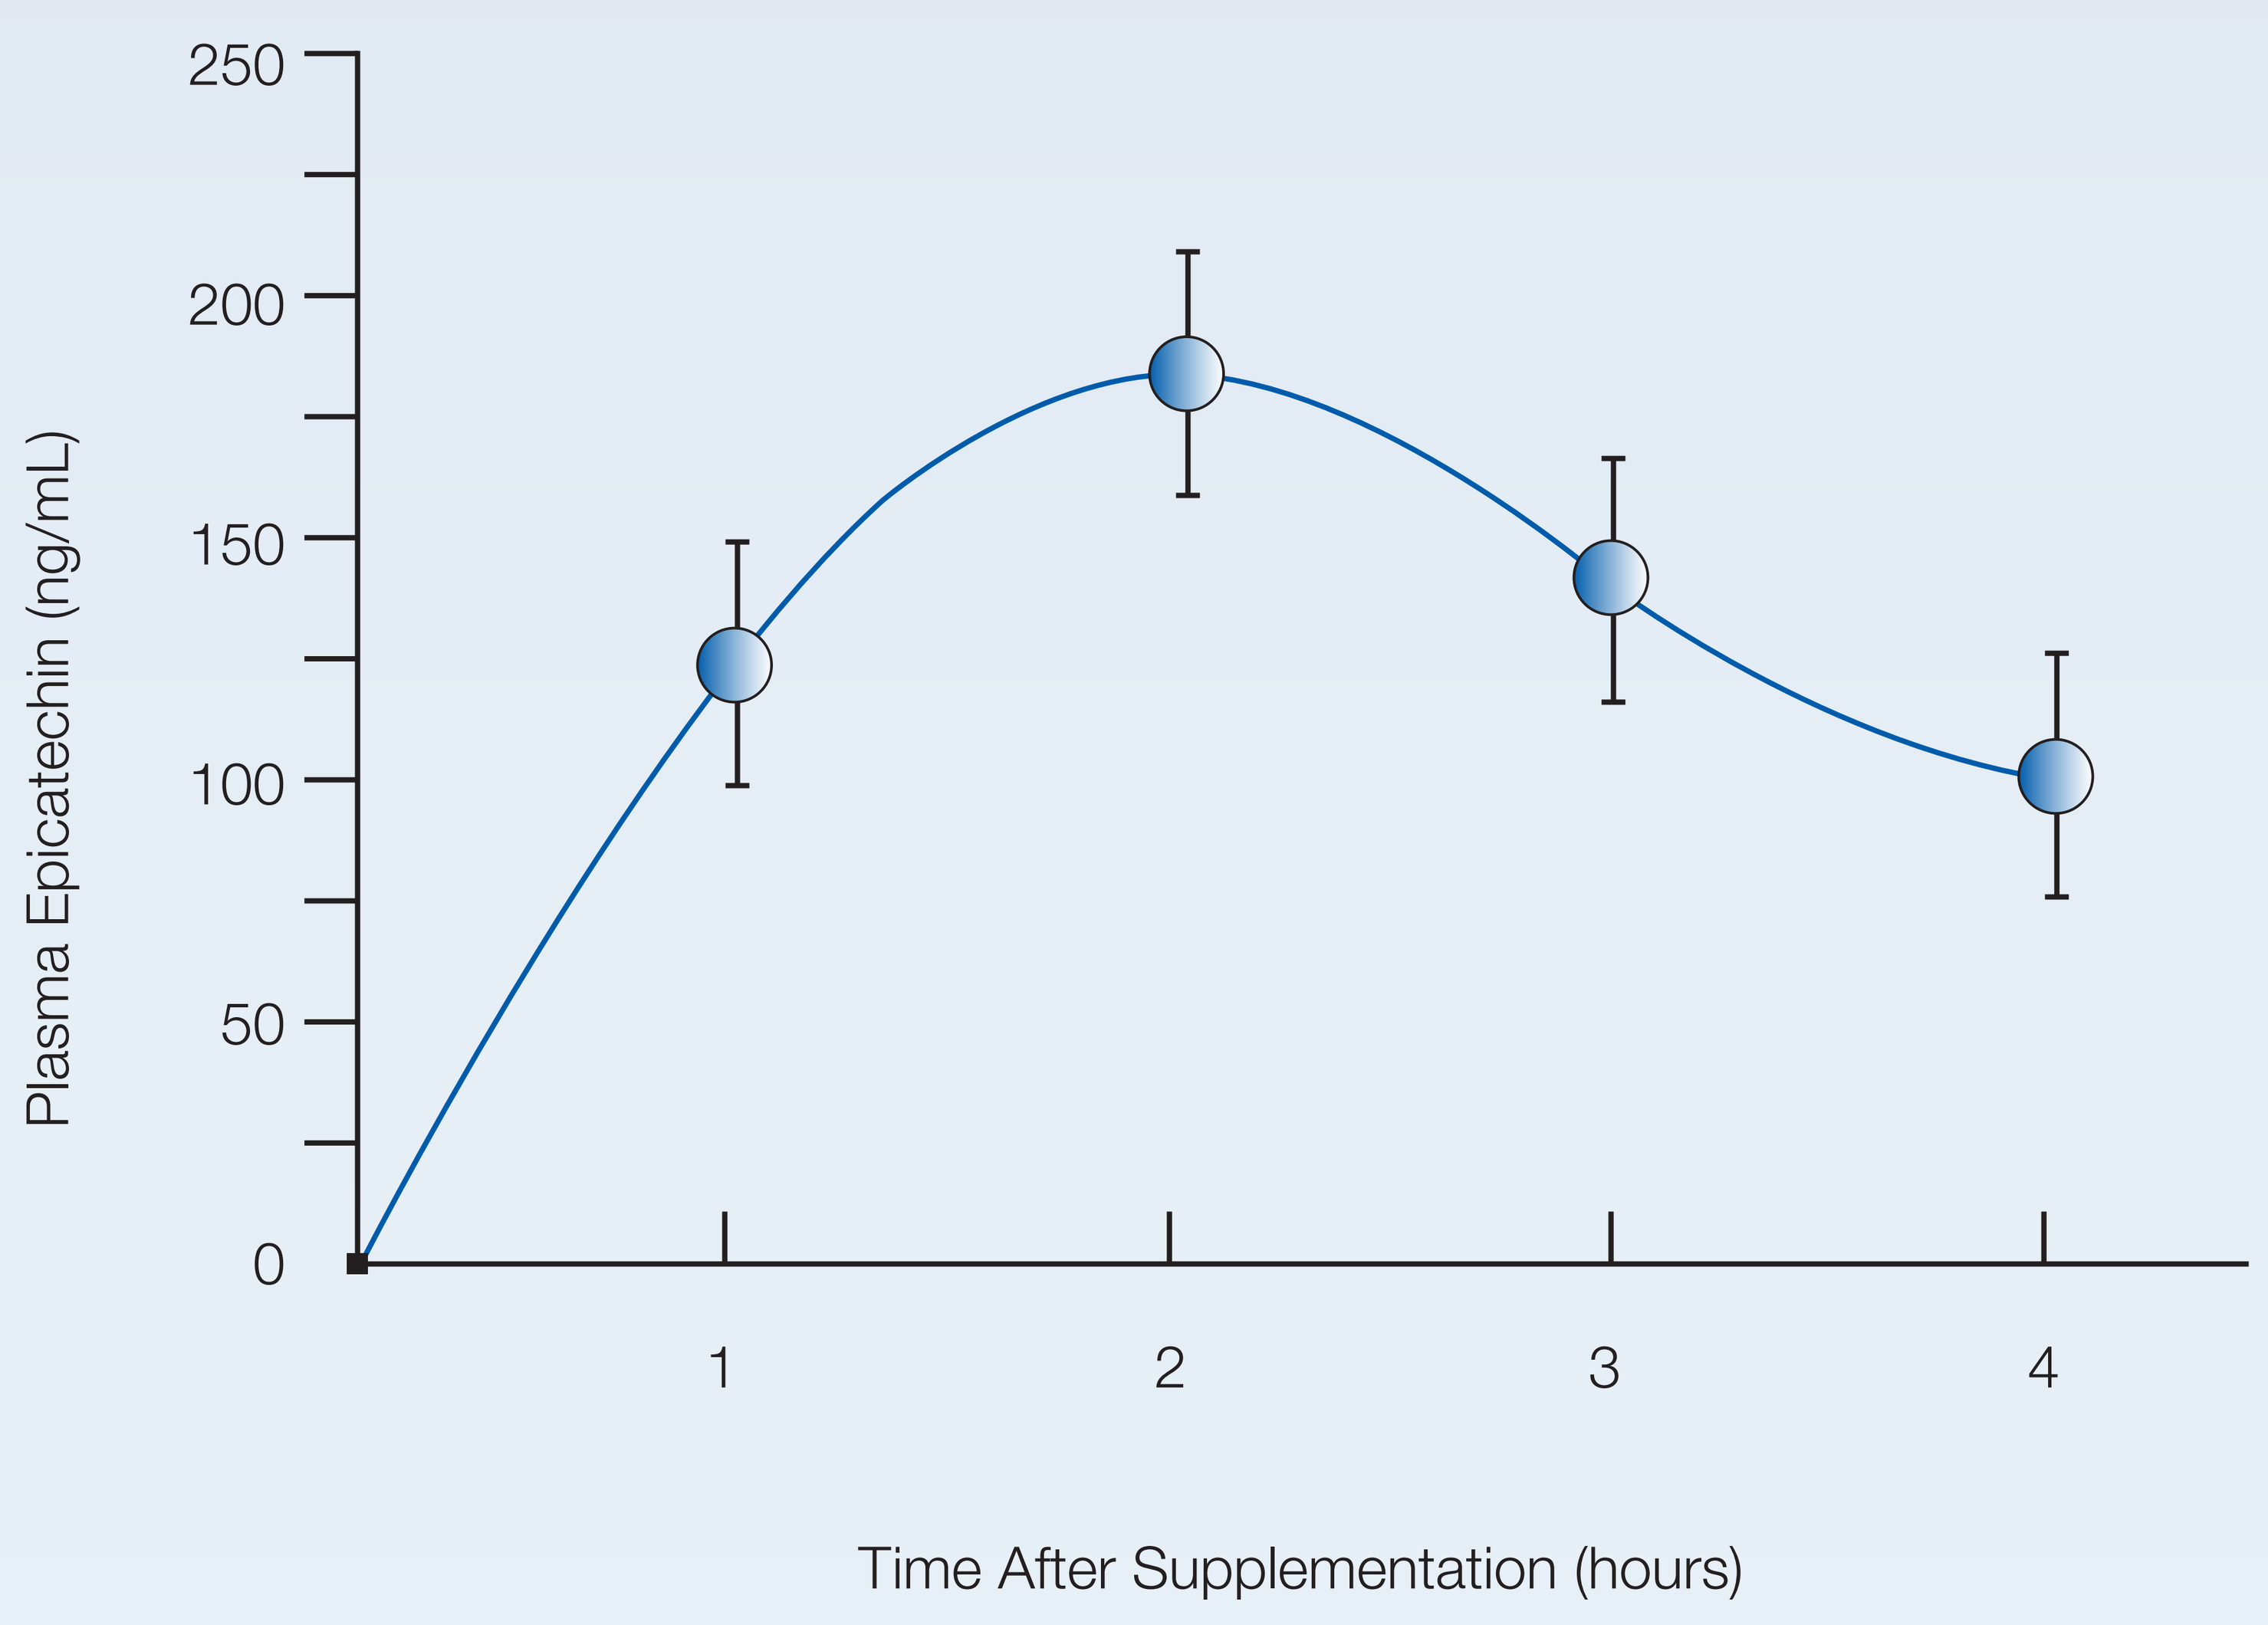
250
200
150
100
50
0
Plasma Epicatechin (ng/mL)
1
2
3
4
Time After Supplementation (hours)

Figure 1. Time course of epicatechin in human plasma as measured by LC-MS after supplementation of 1 g grape seed extract. Error bars are ± standard error of mean.

Grape Seed Extract Increases Plasma Antioxidant Reserve

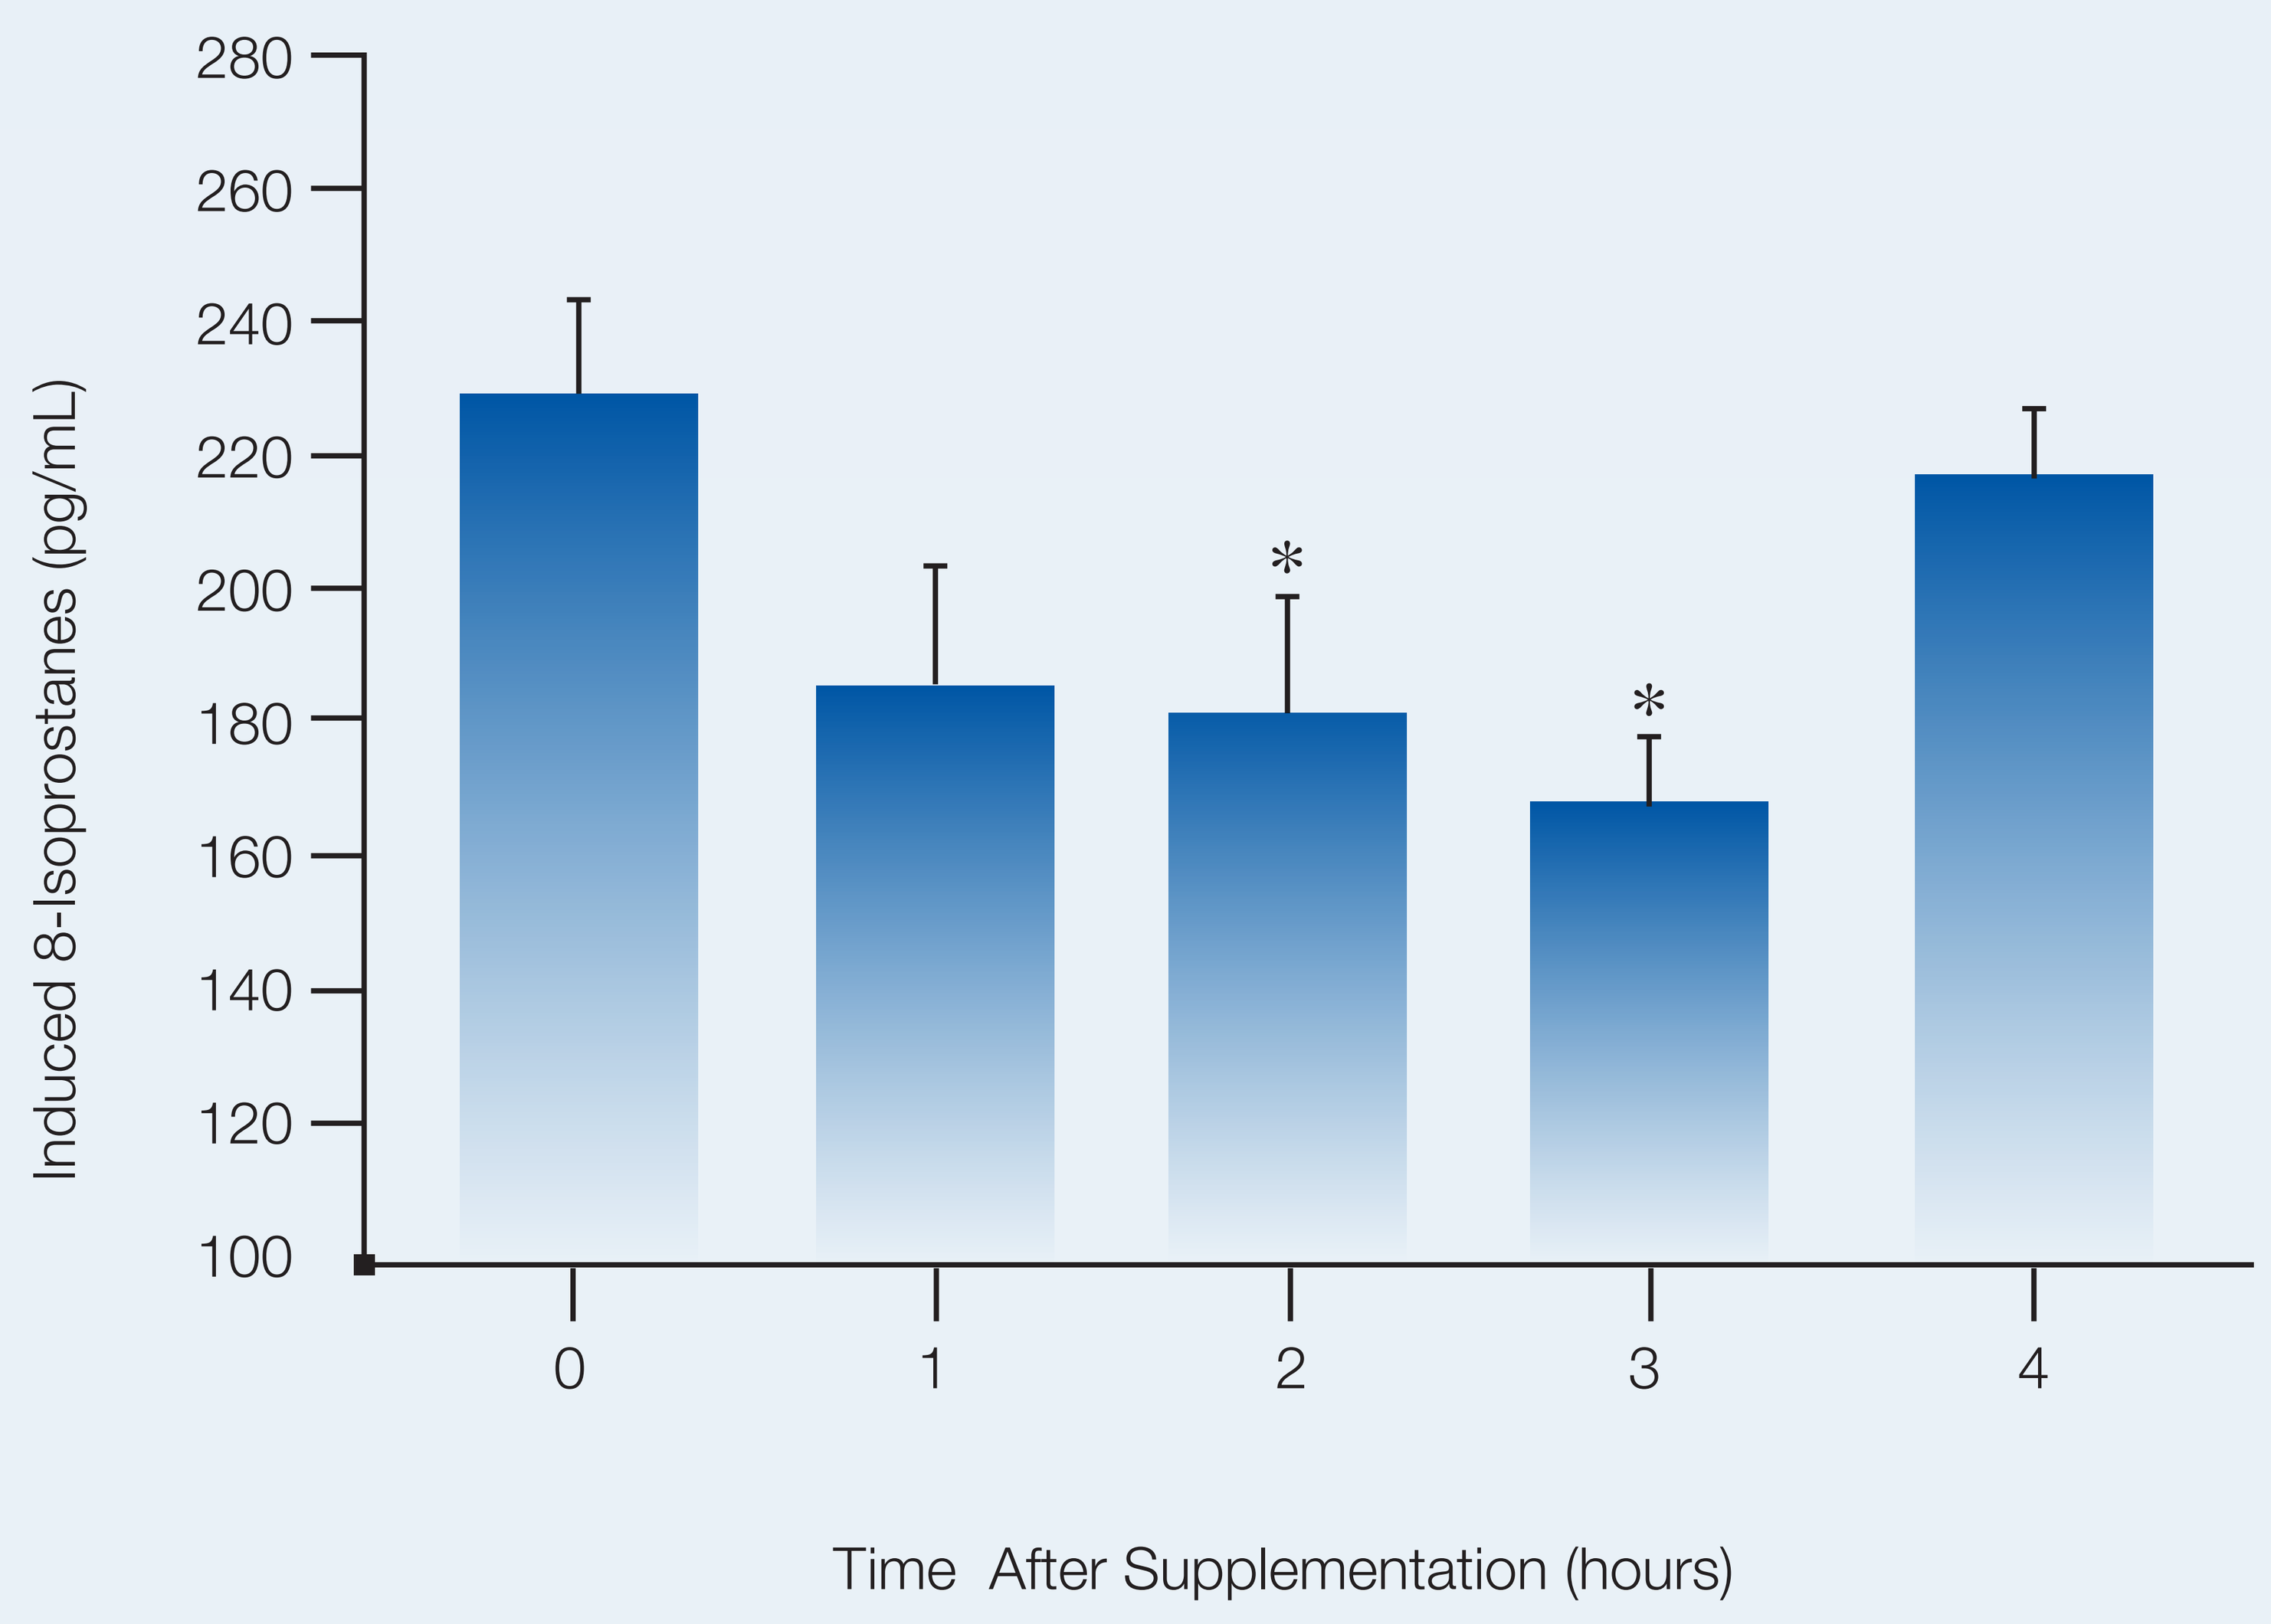
280
260
240
220
200
180
160
140
120
100
Induced 8-Isoprostanes (pg/mL)
*
*
0
1
2
3
4
Time After Supplementation (hours)

Figure 2. Induced 8-isoprostane formation after supplementation with 1 g grape seed extract. Error bars are ± standard error of mean. P values were calculated from absolute values at baseline and 2 h and 3 h after supplementation by a paired t-test. *P<0.05.

Mass Spectrometry

Agilent 1100 series MSD single quadrupole instrument equipped with an ES source was used for this investigation. The nebulizer gas (nitrogen, 99.5% purity) and the drying gas (nitrogen, 99.5%, 350°C) were set to 40 psi and 12 L/min, respectively. The fragmentor voltage was set to 100 V. LC-MS determinations were performed by operating the MSD in the negative ion mode using m/z 289.3.

Preparation of Samples

Plasma samples for HPLC were prepared according to an adaptation of a method first reported by Bell, et al[1]. In brief, samples were treated with β-glucuronidase and then diluted with $CaCl_2$. Samples were incubated at 37°C for 45 min. The samples were then extracted with ethyl-acetate and centrifuged for 10 min at 3000 rpm. The ethyl-acetate layer was then transferred into a cylindrical glass flask and evaporated to dryness at 40°C. Samples were reconstituted in methanol and analyzed via LC-MS.

Plasma Antioxidant Reserve

Baseline, 1, 2, 3 and 4 h plasma samples from each subject were treated with an enzyme mixture of catalase (50 U/mL) & uricase (2.5 U/mL) in 0.15 M NaCl and incubated at 25°C for 10 min. Samples were then treated with SIN-1 chloride (0.2 mmol) and were incubated at 37°C for 4 h with shaking. Plasma 8-isoprostane: were measured following incubation with SIN-1 using an ELISA kit (8-iso Prostaglandin F2 Kit, Cayman Chemical, Ann Arbor, MI). Calibration, curve fitting, and data analysis were done according to the instructions of the manufacturer.

RESULTS

No measurable amount of epicatechin was detected in the samples obtained prior to GSE supplementation (limit of detection is 1 ng/mL). Within the first hour post-supplementation, plasma EC levels began to rise to a concentration of 130 ng/ml. Plasma epicatechin concentration had a maximum of 172 ng/mL by 2 h after ingestion (P <0.001). However, there is no discernable difference between the 1, 2 and 3 h samples and thus should be reviewed carefully before determining an absolute maximum (P <0.13). EC began to decrease by the 3 and 4 h plasma samples, but had not yet returned to baseline concentration (Figure 1).

Within one hour, an increase in plasma antioxidant reserve was seen (P <0.065). At 2 and 3 h there were significant 21% and 27% increases in PAR over the baseline mean (P <0.04 and 0.02, respectively) (Figure 2).

DISCUSSION

Our results demonstrate that epicatechin can be measured via LC-MS in plasma following supplementation of grape seed extract. Furthermore, the data also support the concept that supplementation of grape seed extract can increase total plasma antioxidant capacity as seen using the PAR assay. Consistent with the antioxidant properties of GSE, there were more isoprostanes formed in the baseline sample than in any of the samples taken after ingesting grape seed extract showing that the ingested antioxidants, in fact, can improve the resistance of plasma to oxidation.

[1] J. R. C. Bell, et al. (2000) (+)-Catechin in human plasma after ingestion of a single serving of reconstituted red wine. The American Journal of Clinical Nutrition, 71, 103-108.

[2] A. Rabovsky, J. Cuomo and N. Eich (2006) Measurement of plasma antioxidant reserve after supplementation with various antioxidants in healthy subjects. Clin.Chim.Acta, 371, 55-60.

USANA HEALTH SCIENCES